Determination of an Optimal Formulation for CAR T–Cells: Cryopreservation Studies using Model T-Cells

Rachel Witts[1], Michael Tennant[2], Ken Chrobak[3], Parag Kolhe[1]

[1]*Pharmaceutical R&D, Biotherapeutics Pharm. Sci., Pfizer Inc., Chesterfield, MO*
[2]*Bioprocess R&D, Biotherapeutics Pharm. Sci., Pfizer Inc., Chesterfield, MO*
[3]*BTxPS Tech & Innov, Biotherapeutics Pharm. Sci., Pfizer Inc., South San Francisco, CA*

ABSTRACT

Formulation development for T-cells is challenging due to various considerations including optimization of the hold times during filling and after thawing for CART cell administration. Optimization of the formulation of a cryopreservation medium for CART cells was determined through small and large scale formulation screens utilizing a model T-cell line. Pan T-cells were formulated in various combinations of harvest medium and cryopreservation medium to evaluate if any offered superior cell viability and viable cell density (VCD) following thaw when compared to the platform formulation containing HSA and high DMSO. Viability and VCD were measured immediately after thaw and up to 3 days post reconstitution to determine any effect of the cryopreservation media on the cell recovery. Initial screens showed that a difference between cryopreservation media could be observed and larger formulation robustness studies confirmed this difference. These formulations were further evaluated in CART cells. The results of these studies confirm the development of a robust cryopreservation medium for T-cell formulation.

Initial Formulation Screens

Comparison of Cryopreservation Media

- The platform cryopreservation medium (Saline with 8%HSA and 15% DMSO; final DMSO concentration 7.5%) was compared to CryoStor10 (10% DMSO; final DMSO concentration 5%). All cells were suspended in saline and formulated to 50E6 cells/mL

Figure 1. Formulation and Reconstitution Process

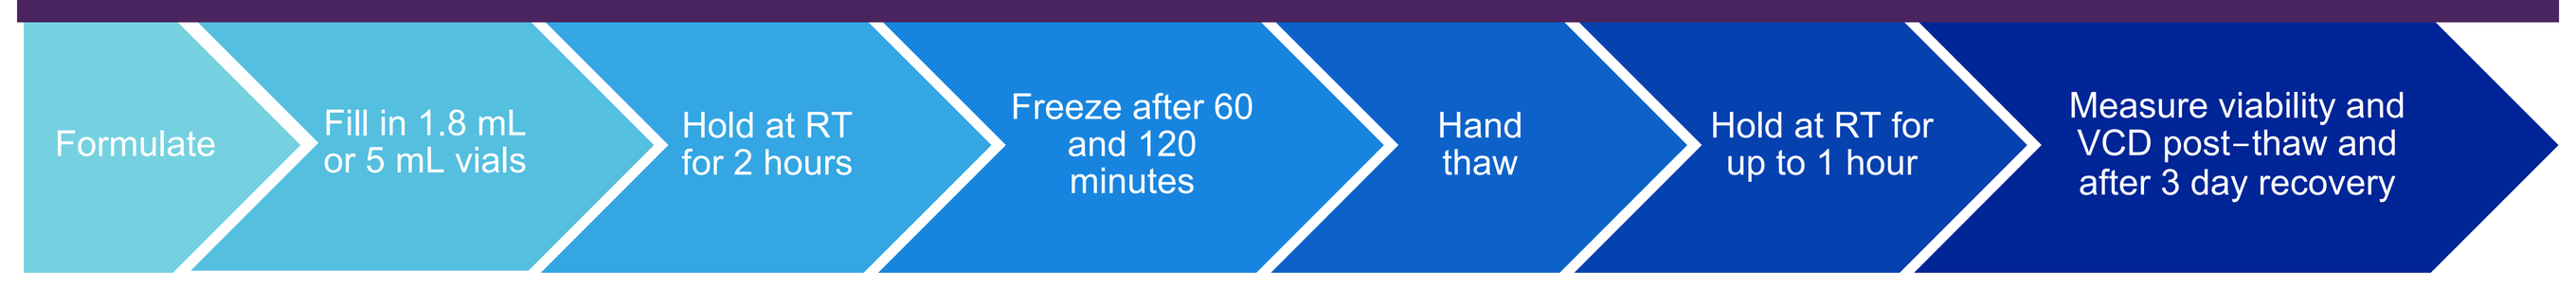

Formulate
Fill in 1.8 mL or 5 mL vials
Hold at RT for 2 hours
Freeze after 60 and 120 minutes
Hand thaw
Hold at RT for up to 1 hour
Measure viability and VCD post-thaw and after 3 day recovery

Figure 2. Average Viable Cell Density by Formulation in 5 mL Vials After Three Days of Recovery Post Thaw – 7.5% DMSO versus CS10

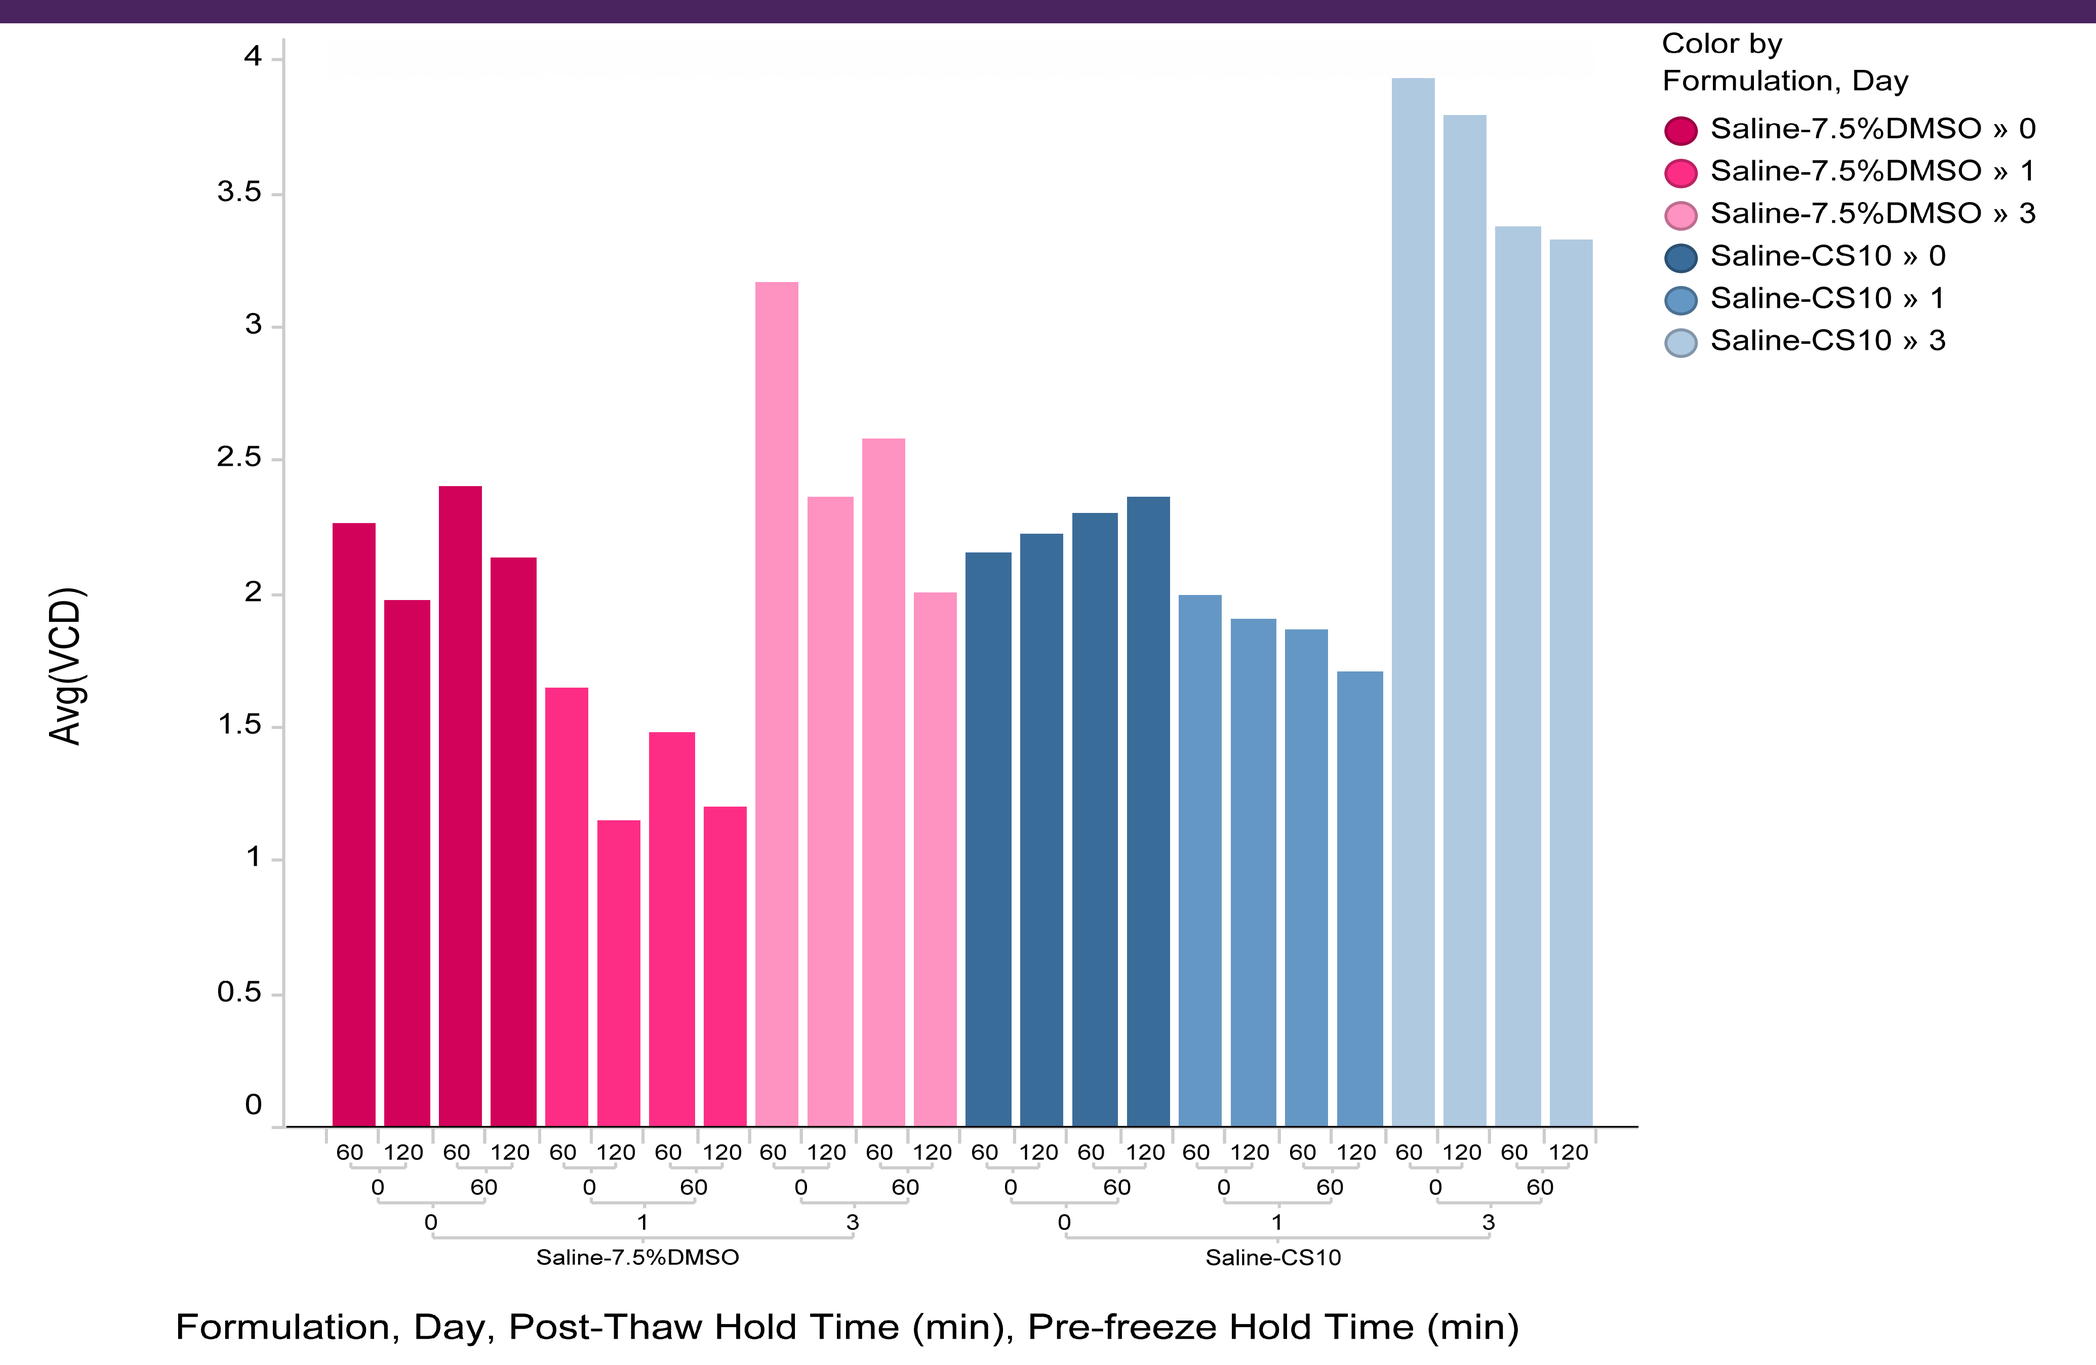

Avg(VCD)
Color by Formulation, Day
Saline-7.5%DMSO » 0
Saline-7.5%DMSO » 1
Saline-7.5%DMSO » 3
Saline-CS10 » 0
Saline-CS10 » 1
Saline-CS10 » 3
Saline-7.5%DMSO
Saline-CS10
Formulation, Day, Post-Thaw Hold Time (min), Pre-freeze Hold Time (min)

Cells formulated in CryoStor10 (CS10) showed better recovery than cells formulated in the plaftorm formulation.

Comparison of Harvest Media

- To determine if harvesting in Cryostor Basal (CSB) medium would provide added cryopreservative effects over saline. Cells were formulated to 50E6 cells/mL.

Figure 3. Average Viable Cell Density by Formulation in 5 mL vials after Three Days of Recovery Post Thaw (2 Runs) – Saline versus CSB

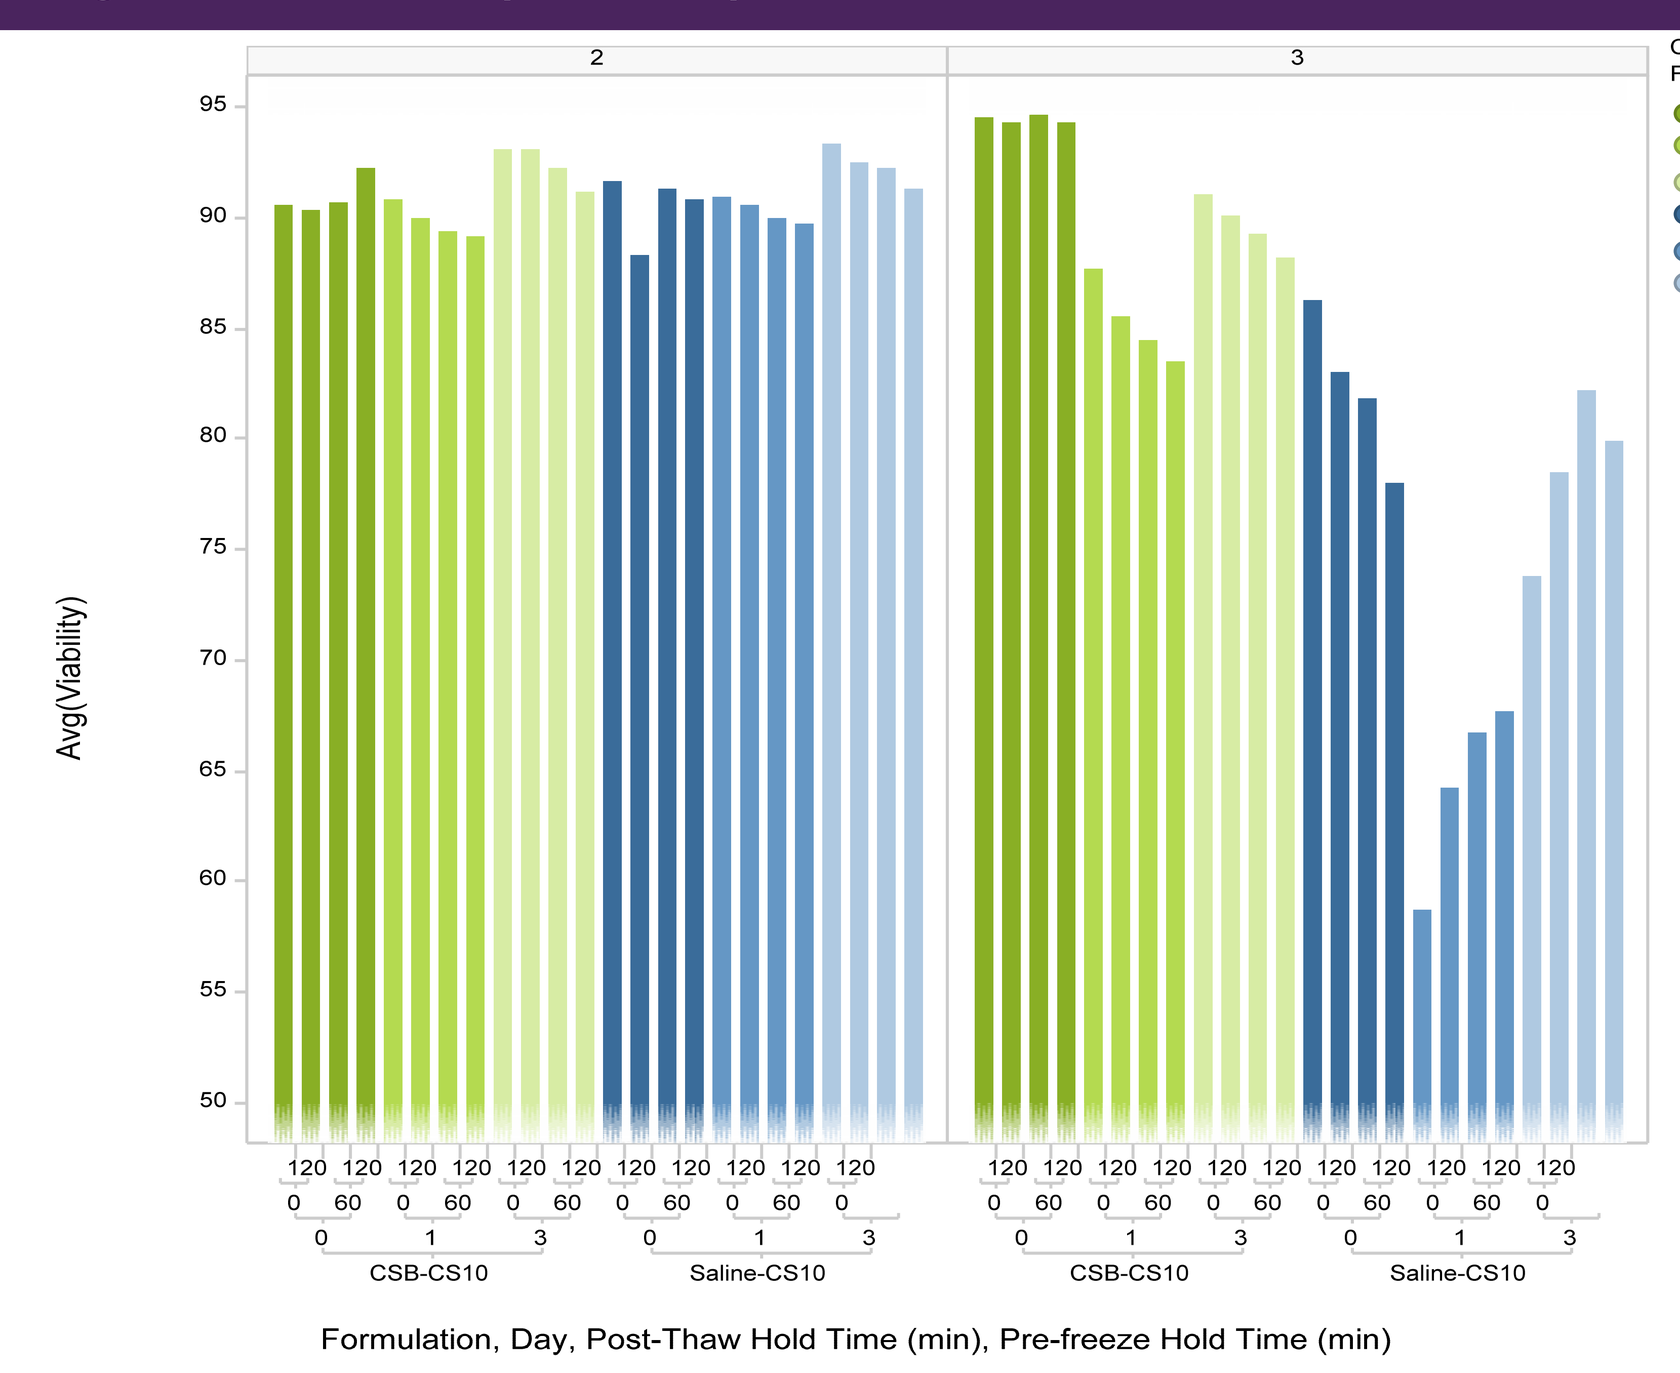

2
3
Avg(Viability)
Color by Formulation, Day
CSB-CS10 » 0
CSB-CS10 » 1
CSB-CS10 » 3
Saline-CS10 » 0
Saline-CS10 » 1
Saline-CS10 » 3
CSB-CS10
Saline-CS10
Formulation, Day, Post-Thaw Hold Time (min), Pre-freeze Hold Time (min)

CryoStor Basal media may provide some added cryopreservative effects over saline.

Formulation Robustness Study

- Cells were formulated, filled into 1.8 mL vials at 50E6 cells/mL and held at room temperature for up to 3 hours. Cells were frozen after 120 minutes and 180 minutes and held on stability in $vPLN_2$ for up to 3 months.

Table 1. Formulation Descriptions

Formulation ID	Basal medium	2X cryopreservation medium	Final DMSO concentration
F1 (platform)	Saline (0.9%)	PBS, 8%HSA, 15% DMSO	7.5% DMSO
F2	Saline (0.9%)	CryoStor10	5% DMSO
F3	CryoStor CSB	CryoStor10	5% DMSO
F4	CryoStor CSB	PBS, 8%HSA, 10% DMSO	5% DMSO

- Following 37°C waterbath thaw, cells were held at 4°C, 22°C, and 37°C for up to 2 hours. Viability and VCD were measured post-thaw and after 3 days of recovery.

Figure 4. Average Viability by Formulation After Three Days of Recovery Post Thaw (T0 data)

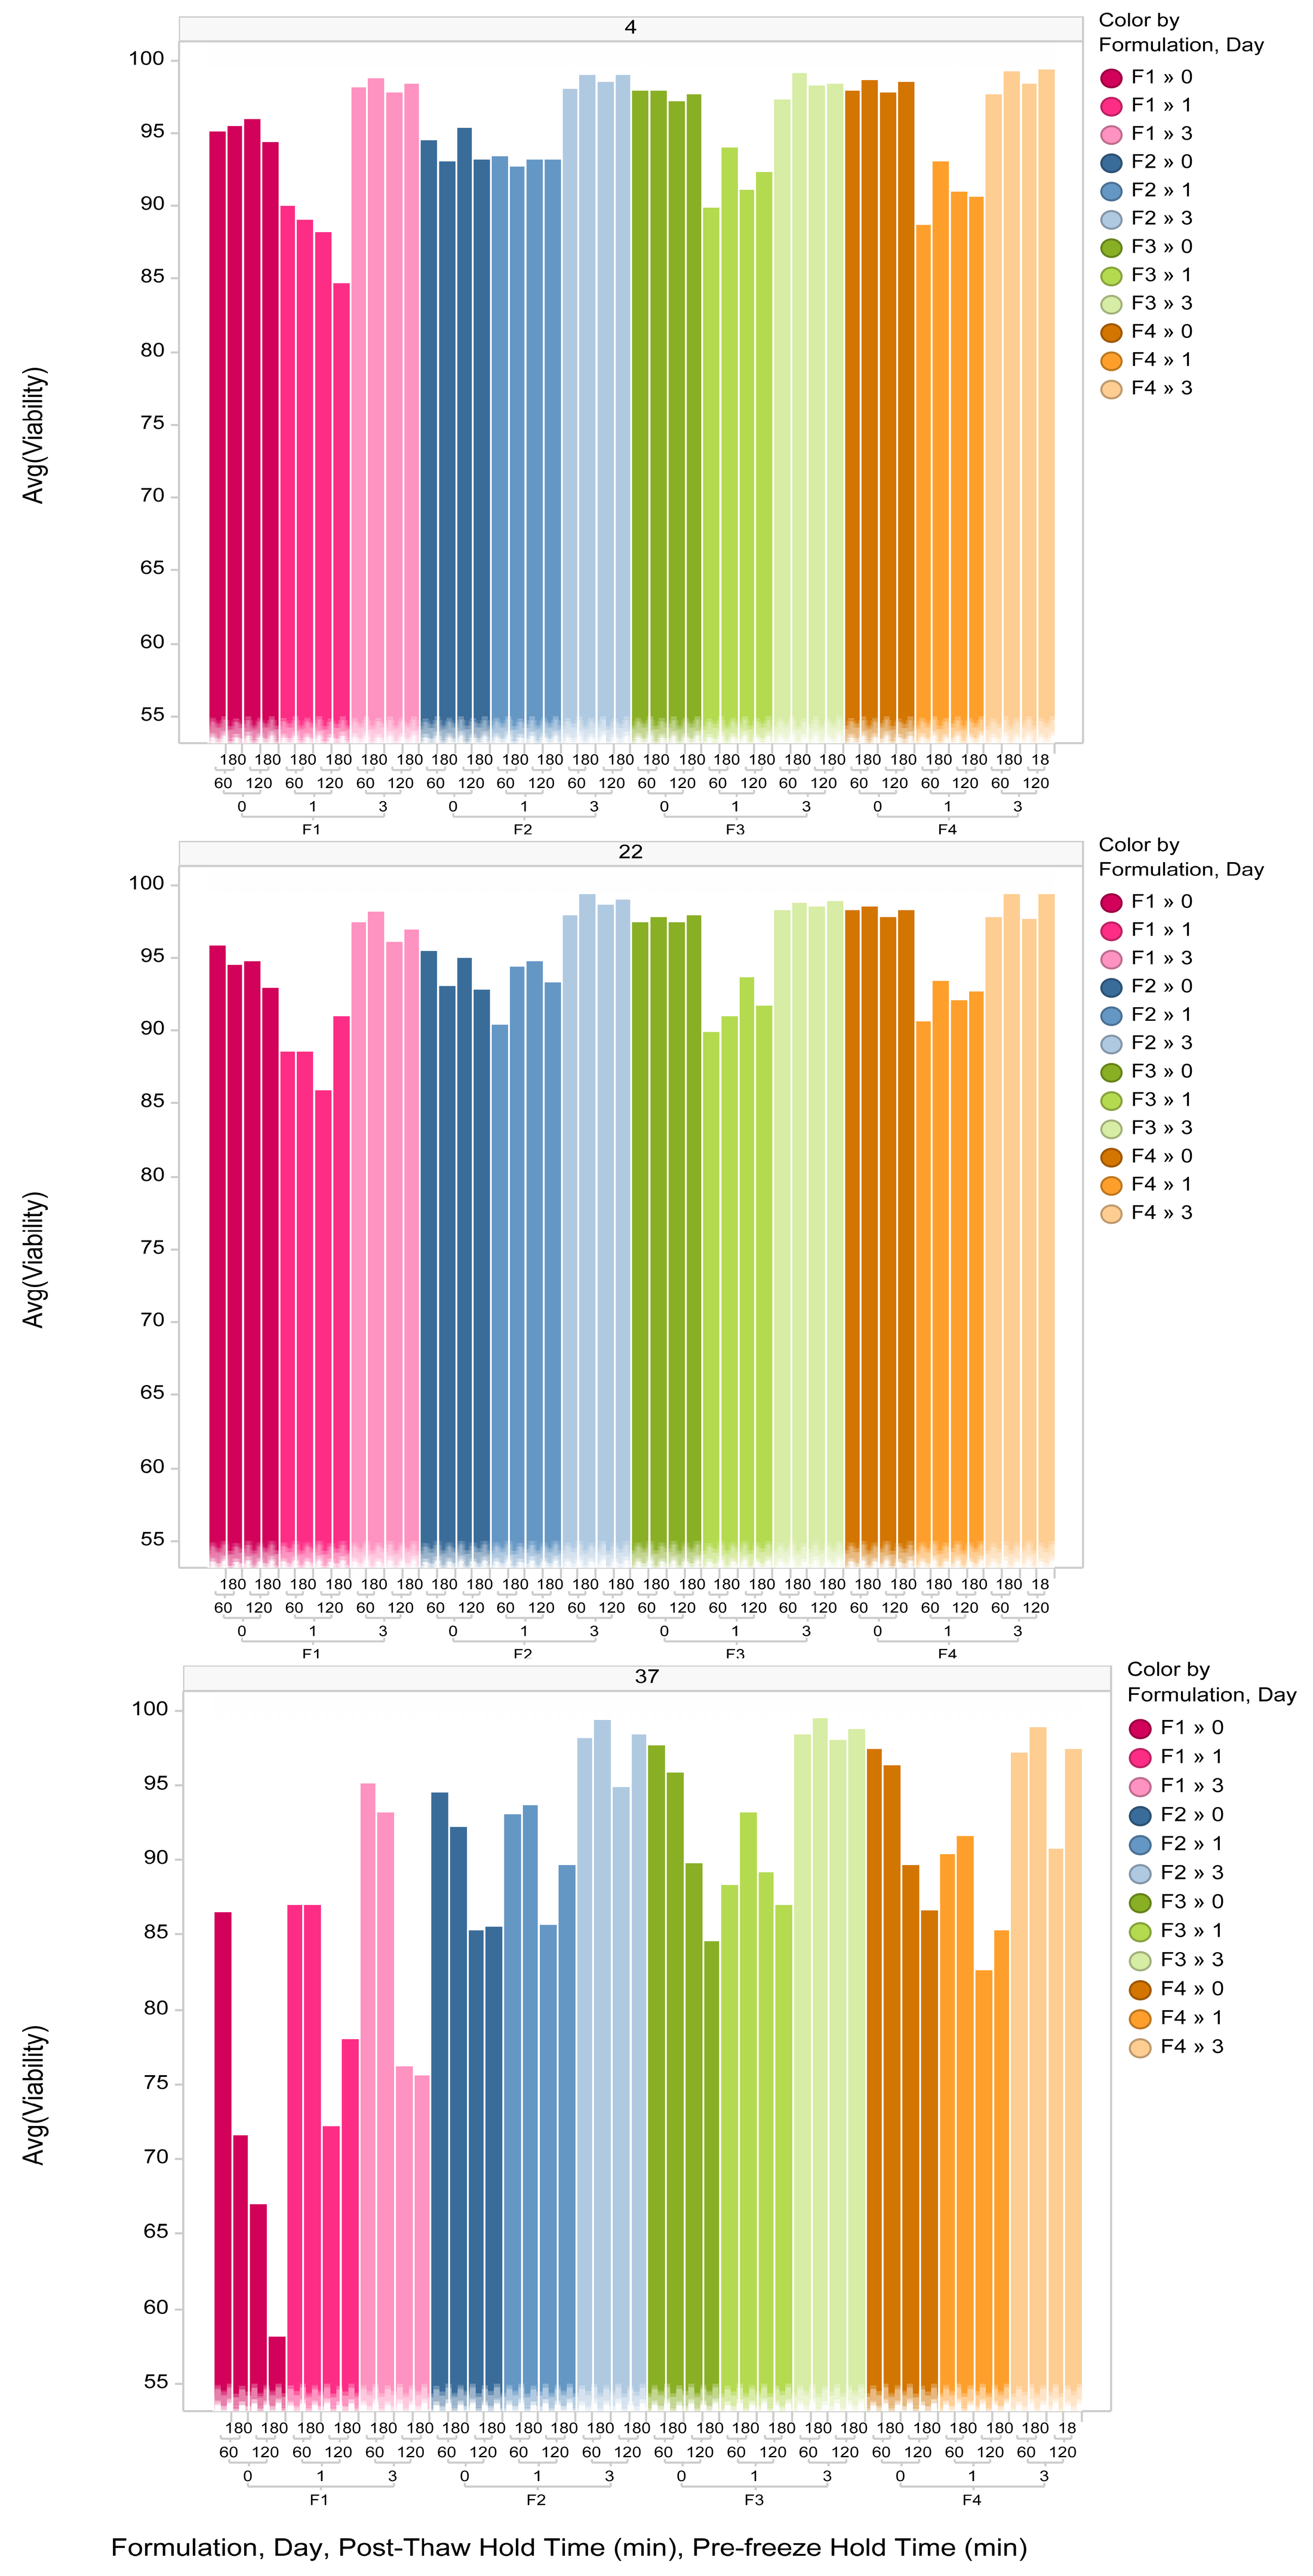

4
22
37
Avg(Viability)
Color by Formulation, Day
F1 » 0
F1 » 1
F1 » 3
F2 » 0
F2 » 1
F2 » 3
F3 » 0
F3 » 1
F3 » 3
F4 » 0
F4 » 1
F4 » 3
Formulation, Day, Post-Thaw Hold Time (min), Pre-freeze Hold Time (min)

The platform formulation showed decreased viability and recoverable TVC (data not shown) at 37°C compared to the other formulations.

CART Cell Evaluation

- CAR positive T-cells were formulated in the platform formulation or in CS10 with saline as the suspension reagent and formulated to >50E60 cells/mL in 2 mL vials. Cells were held pre-fill up to 120 minutes and pre-freeze up to an additional 120 minutes. Post thaw, cells were allowed to recover over 2 days.

Figure 5. Average Viability by Formulation Post Thaw – CART cells

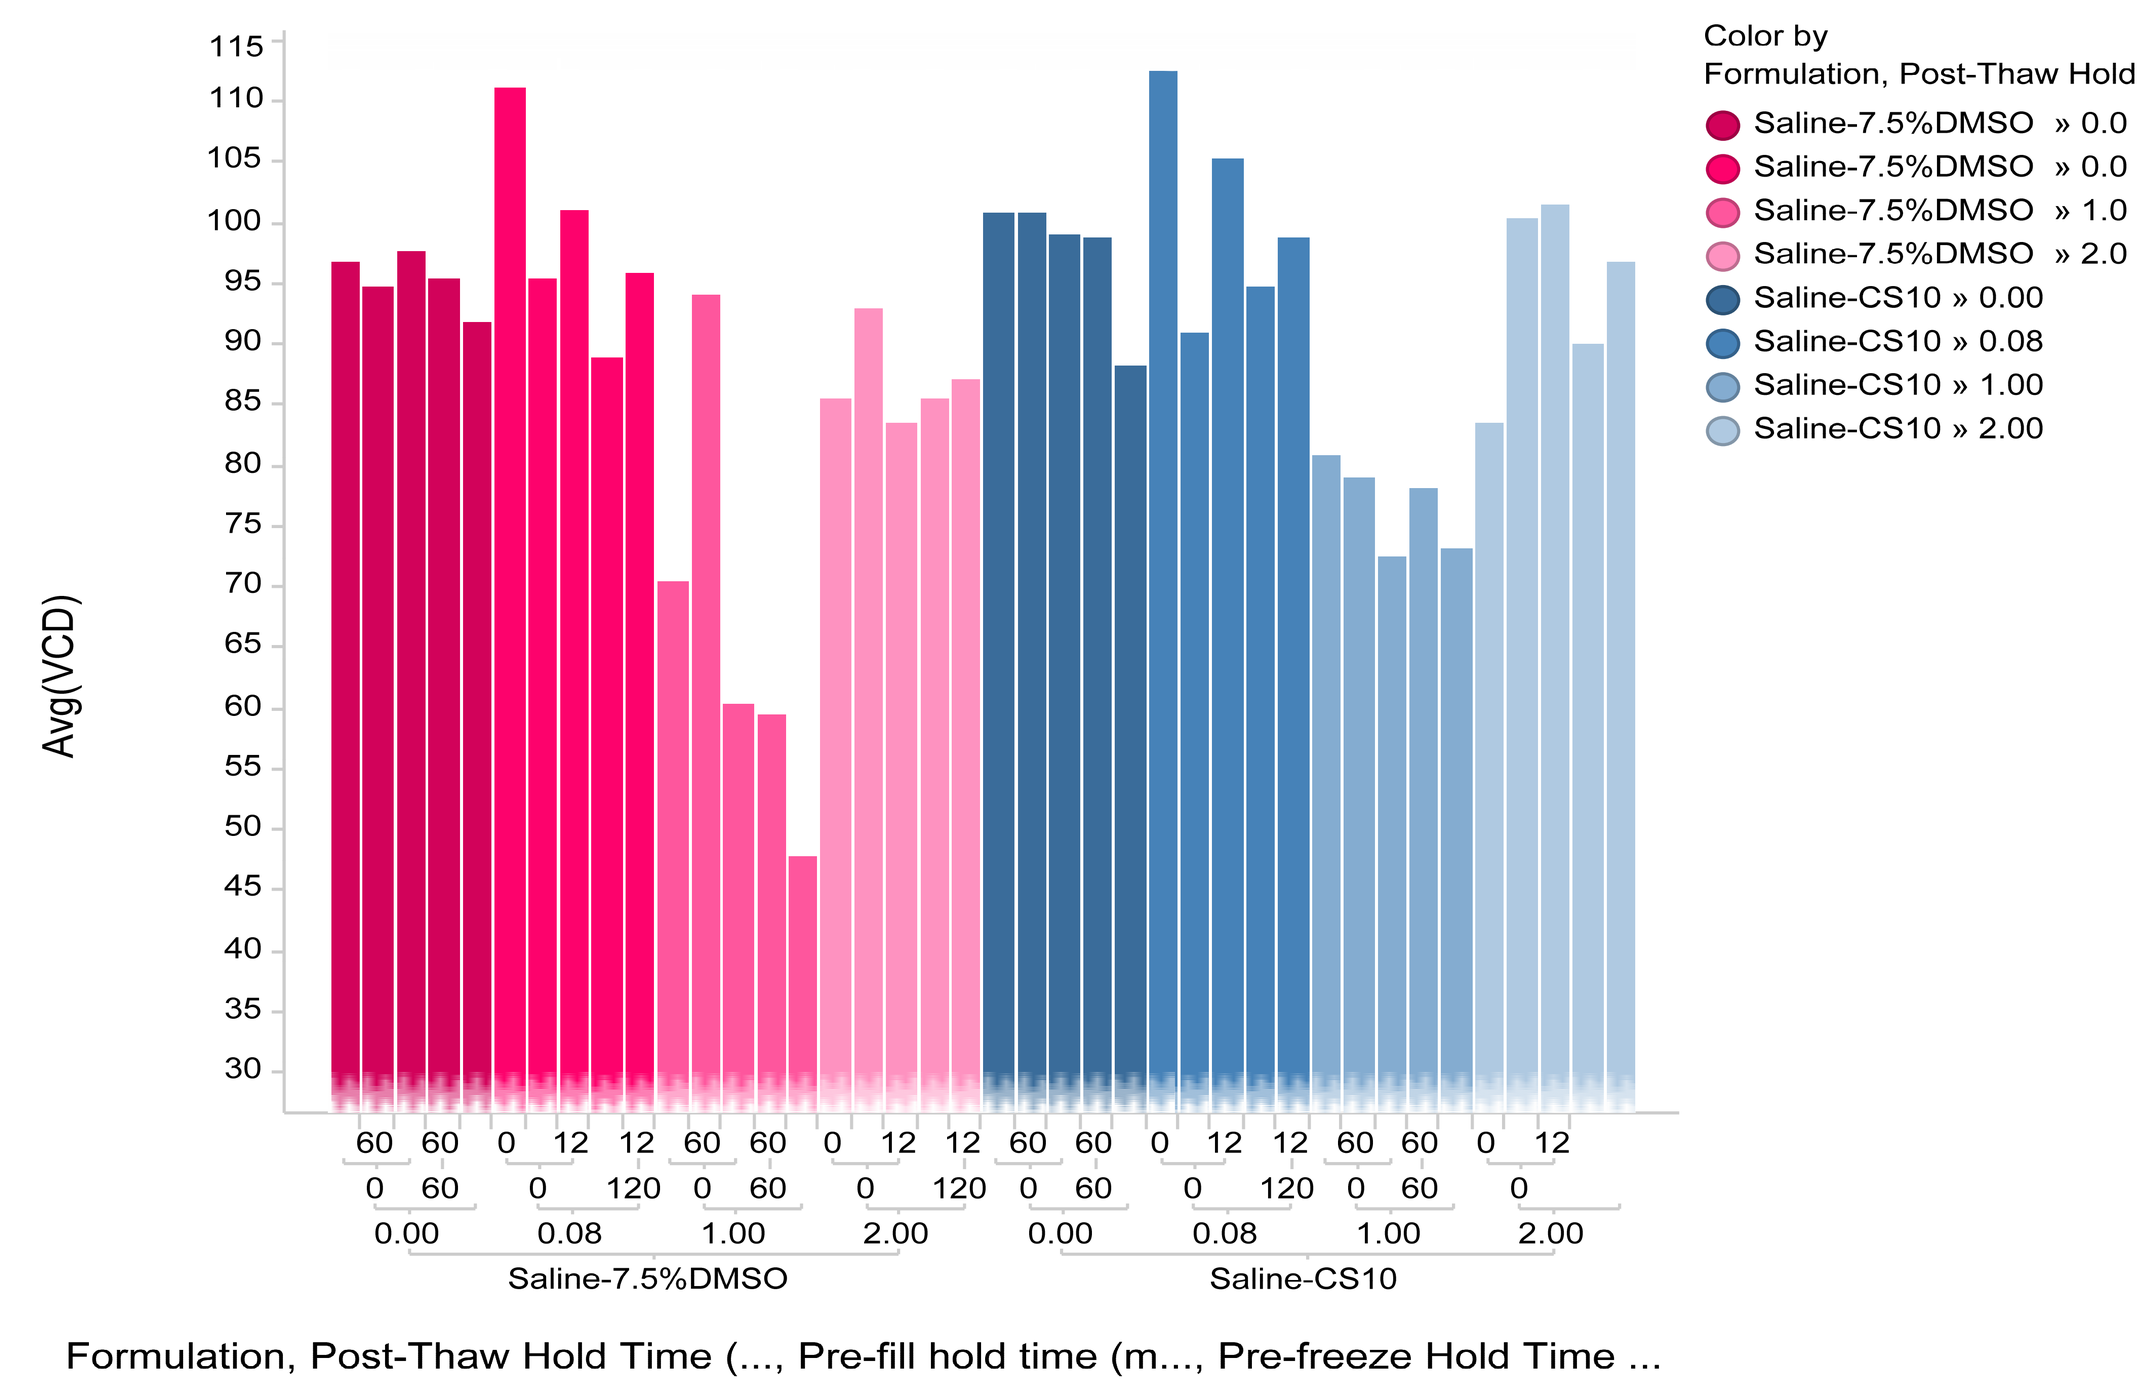

Avg(VCD)
Color by Formulation, Post-Thaw Hold
Saline-7.5%DMSO » 0.0
Saline-7.5%DMSO » 0.0
Saline-7.5%DMSO » 1.0
Saline-7.5%DMSO » 2.0
Saline-CS10 » 0.00
Saline-CS10 » 0.08
Saline-CS10 » 1.00
Saline-CS10 » 2.00
Saline-7.5%DMSO
Saline-CS10
Formulation, Post-Thaw Hold Time (..., Pre-fill hold time (m..., Pre-freeze Hold Time ...

- Cells were also formulated in CS10 with CSB as the suspension reagent and are compared with Pan T-cells in the same formualtion.

Figure 6. Average Viability by Formulation Post Thaw CART cells versus Pan T-cells

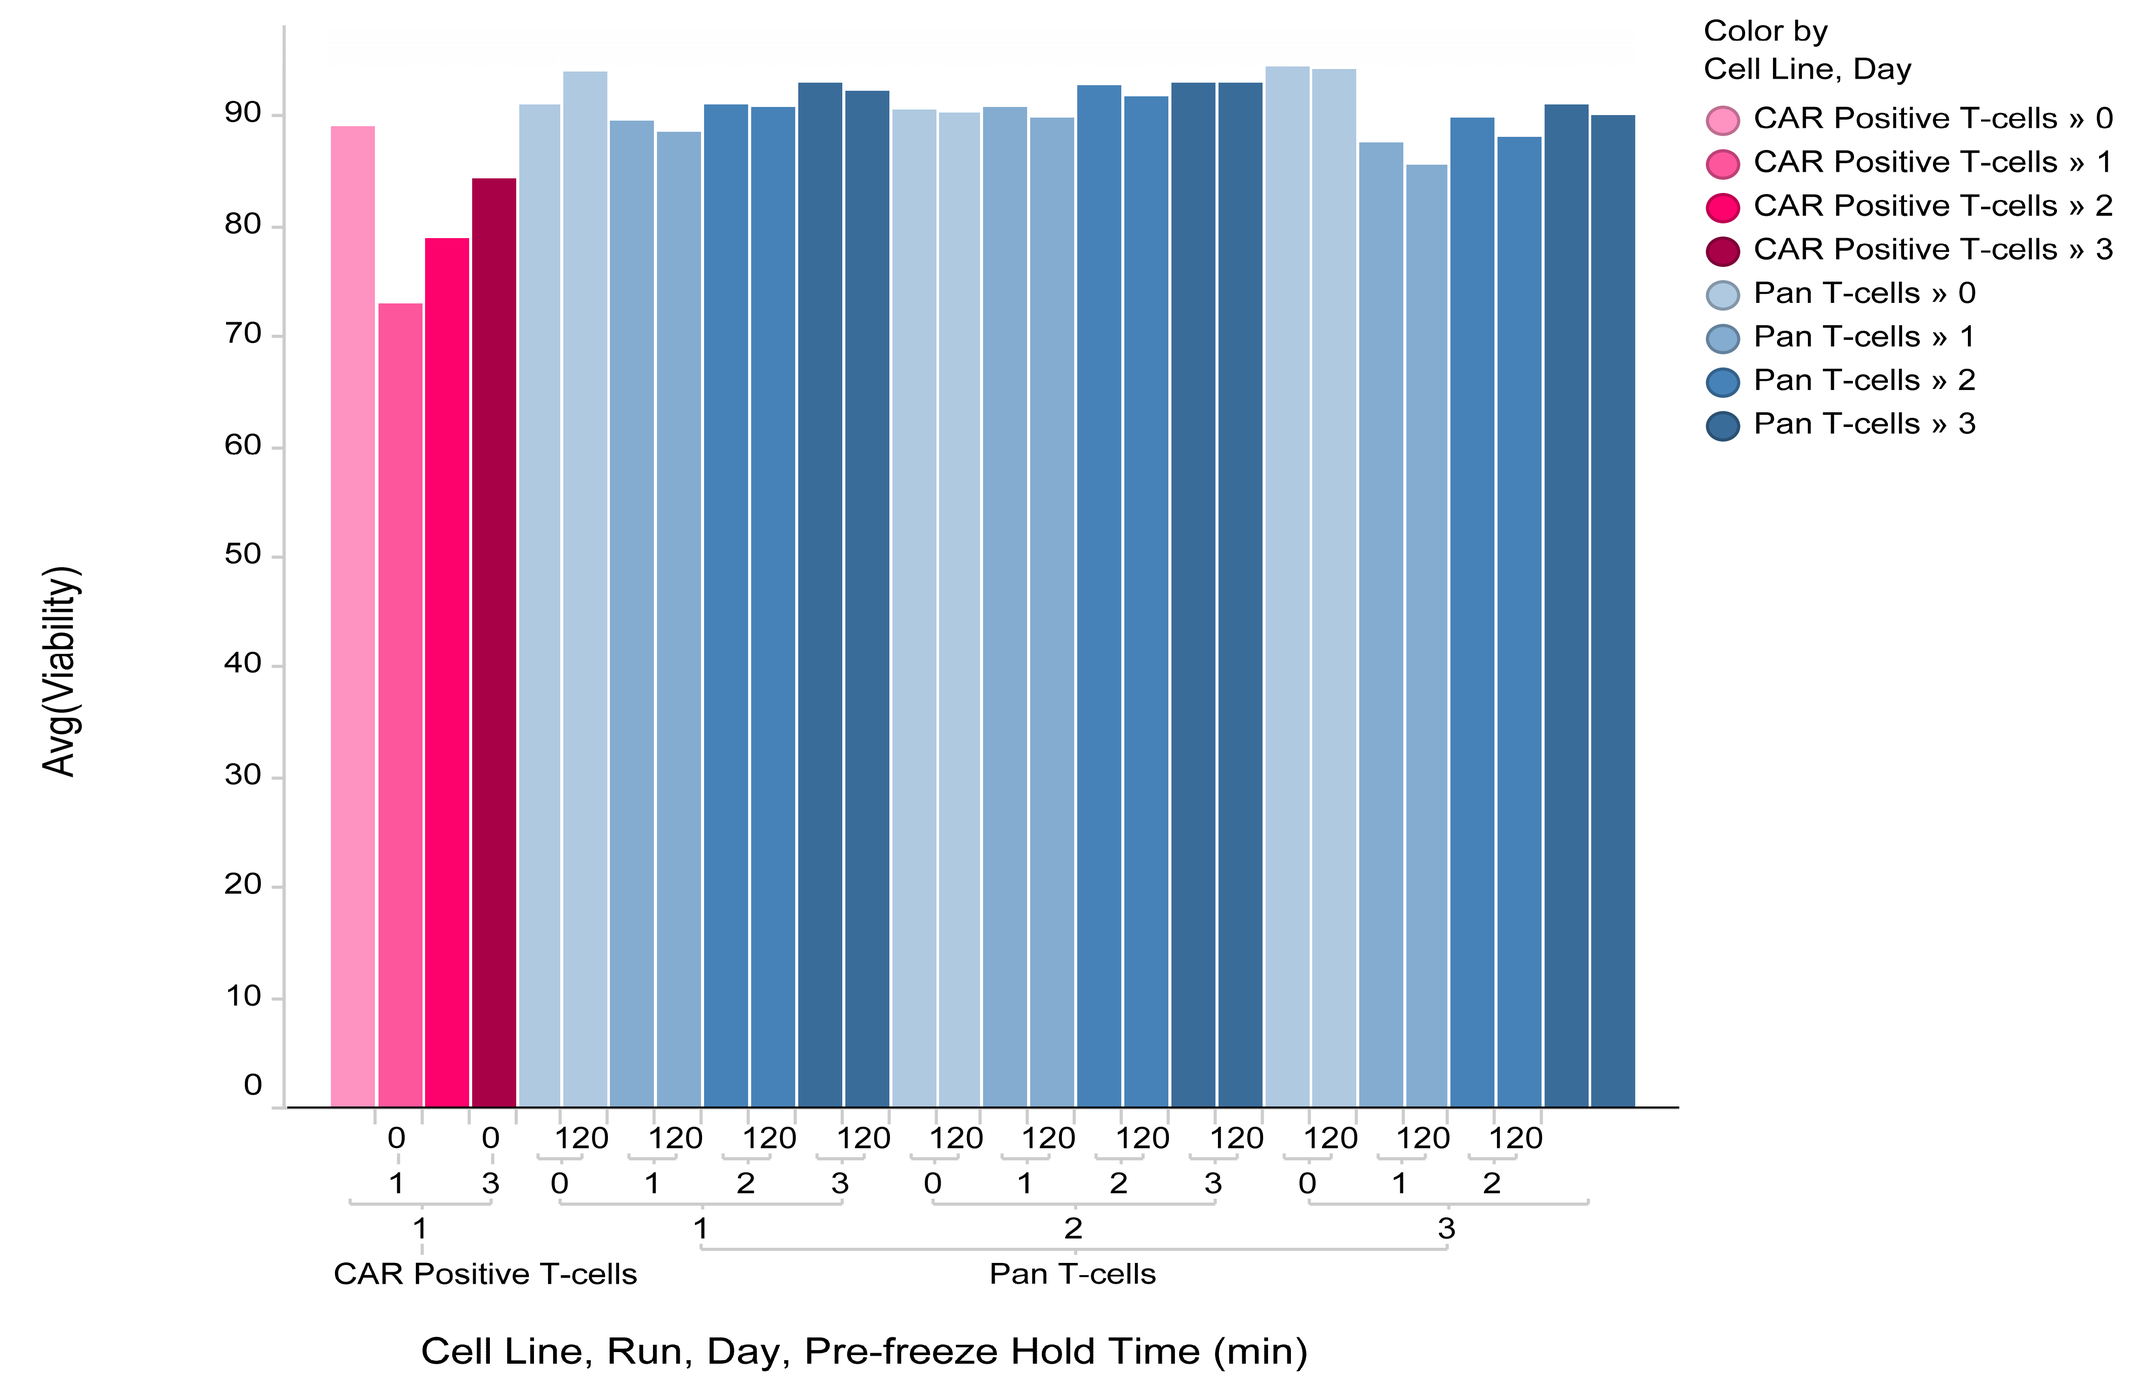

Avg(Viability)
Color by Cell Line, Day
CAR Positive T-cells » 0
CAR Positive T-cells » 1
CAR Positive T-cells » 2
CAR Positive T-cells » 3
Pan T-cells » 0
Pan T-cells » 1
Pan T-cells » 2
Pan T-cells » 3
CAR Positive T-cells
Pan T-cells
Cell Line, Run, Day, Pre-freeze Hold Time (min)

Using a formulation with CS10 gives comparable results to the platform formulation. Additionally, comparable results were obtained from cells formulated in CSB + CS10 when observed with CART cells.

CONCLUSIONS

The data from the inital formulation screens showed that a discernible difference between formulations could be observed. Pan T-cells formulated in CryoStor 10 showed better recovery than cells formulated in the platform formulation after 3 days and suspending cells in CSB rather than saline may provide a slight improvement in recovery from cryopreservation.

The large scale formulation study confirmed the platform formulation performed worse than the other formulations tested. There was little difference between the results of post-thaw hold at 4°C and at 22°C, but post-thaw hold at 37°C showed a decrease in viability and VCD for the platform formulation.

Examination of the formulations on CART Cells showed that using CS10 in the formulations gave equal viabilities and VCD. This allows for the possibility of moving to a formulation without HSA. Additionally when compared to Pan T-cell data, there was not a significant difference when formulated in CS10 with CSB. In pan T-cells, clumping was observed in some Saline containing formulations, suggesting the need to move to a CSB based formulation. Further evaluation will be conducted to determine the final formulation for the CART cells.

ACKNOWLEDGEMENTS

Thanks to the Wuxi collaboration for the formulation robustness data and the entire Cell & Gene Therapy and PharmSci CART team.