Presented at the International Society for Cellular Therapy, North America Meeting
Memphis, TN, October 2016

CONTINUED OUTGROWTH OF CORD TISSUE-DERIVED CELLS OVER MULTIPLE PLATINGS FROM CORD TISSUE CRYOPRESERVED AS A COMPOSITE MATERIAL

Matthew L. Skiles, Katherine S. Brown, Heather L. Brown
Scientific and Medical Affairs, Cbr Systems, Inc., San Bruno, CA

Cbr® From AMAG Pharmaceuticals

BACKGROUND

Umbilical cord tissue (CT) can be easily collected in a non-invasive procedure and is a rich source of mesenchymal stem cells (MSCs) and other progenitor cell populations with potential therapeutic value. A systematic analysis of the reliability and robustness of explant outgrowth and enzymatic digestion of fresh umbilical cord tissue for the purpose of MSCs isolation concluded that explant outgrowth is highly amenable to large-scale biobanking operations.[1] Furthermore, explant outgrowth can be utilized to isolate MSCs from either fresh cord tissue or tissue cryopreserved as a composite, or raw, material. Cryopreservation of CT as a composite tissue maintains native architectural and biochemical properties of the stem cell microenvironment during storage, properties that continue to be maintained if the tissue is explanted for MSC isolation.

In a typical explanting procedure, tissue is placed in cultureware in the presence of a supportive medium until the cells of interest have migrated onto the growth surface. The tissue is then often discarded and the cells are either harvested or allowed to continue in culture. Several studies have shown the feasibility of a repeated explant culture scheme in which explanted tissue is not discarded but instead plated as a new explant to isolate additional primary cell material. However, to our knowledge, such studies have not systematically examined the consistency of cell outgrowth from each individual explant as opposed to CT as a whole unit. Here, we evaluated the use of a repeated CT explant culture for cell isolation with an assay that allows for characterization and quantitative measurement of outgrowth from individual explants.[2]

METHODS

Donated CT (n=4) was collected from consenting mothers, transported to a processing facility, washed, and segmented into small pieces. A portion of each tissue was explanted fresh (prior to cryopreservation) while another portion was cryopreserved and stored at -196°C in a clinical-grade, DMSO-based cryopreservation freeze medium (CryoStor,® BioLife Solutions) for at least 1.5 years then thawed and explanted. For explant cultures, smaller tissue pieces were cut from the fresh or frozen tissue and placed on tissue culture plates in a gridded pattern with 24 explant locations for each plate. After 7 days, the tissue pieces were removed from the plate and the medium exchanged. For fresh platings, the tissue pieces were discarded. For thawed platings, the tissue pieces were transferred at 7 days into the wells of a 24-well plate (1 tissue piece per well) and medium was added (Figure 1). The tissue pieces were then serially re-plated each week in the same manner for a total of 5 thawed platings. For each individual explant culture, cell growth from each tissue piece was observed and assigned a score of 0 to 4 based on cell attachment and extent of cell proliferation (Figure 2).

RESULTS

All fresh and cryopreserved CT units yielded proliferating, plastic-adherent cells with fibroblastic morphology, yielding a 100% rate of success for isolation. Fresh and frozen CT from the same donor were comparable, as evident from the ratio of fresh to frozen ("Thawed, Plate 1") scores (Table 1), similar to what has previously been described.[2] When compared to the average score of fresh platings or the average score of initial thawed platings, the average score of the first replatings was significantly increased. The average score of subsequent replatings did not differ significantly from the average score of fresh or initial thawed platings.

Outgrowth of adherent cells from individual tissue pieces was fairly consistent, as shown in Tables 2 and 3. On average for each plating, from the initial thawed plating through four serial replatings, the average change in score for a single tissue piece was 0.82 (±0.37) points. Omitting the comparison between the initial thawed platings and the first replates, in which the average score differed significantly, the average change in score for a single tissue piece was 0.65 (±0.18) points. The overall average score range for individual tissue pieces was less than 2 points (1.97 ±0.2) and nearly 75% of wells exhibited minimal change in score over five serial platings.

Total cumulative cell yield per CT unit was increased on average by 25 fold through serially replating and harvesting of cells that continued to migrate out of explanted tissue pieces, as shown in Figure 2.

CONCLUSION

Using an enzyme-free approach, we show continuous outgrowth from individual explants of cryopreserved CT over five serial platings. The capacity of explants to generate cell outgrowth did not appear to diminish over multiple replatings. Furthermore, explant score for individual tissue pieces, representative of the degree of cell outgrowth and proliferation, displayed minimal variation. The ability to replate CT explants provides an opportunity to increase cell recovery from a given tissue under processes that are driven by the cells themselves and has the advantage of retaining the natural matrix, providing a stem cell niche in vitro. Also, cryopreserving CT as composite material allows for isolation of MSCs at the point of care when the specific clinical application is known. Harnessing the availability of the natural MSC environment in cryopreserved cord tissue may be particularly attractive for tissue engineering applications, where growth factors, scaffolds and stem cells are each key elements.

REFERENCES

1. Iftimia-Mander A, Hourd P, Dainty R, Thomas RJ. Mesenchymal Stem Cell Isolation from Human Umbilical Cord Tissue: Understanding and Minimizing Variability in Cell Yield for Process Optimization. *Biopreservation and Biobanking*. 2013;11:291-298.
2. Skiles, ML, Brown, KS and Brown HL. Development of a Quantitative Measurement of Mesenchymal Stem Cells Explant Outgrowth from Umbilical Cord Tissue. *Transfusion*. 2015;55:1A–7A.

Figure 1.
Schematic for Serial Replating and Scoring of Tissue Explants

Initial explant cultures from each CT unit consisted of 24 small pieces of tissue in a round culture dish which were then re-plated by transferring each tissue piece to the corresponding well of a fresh 24-well plate (A). After 2 weeks, each culture was visualized and assigned a score from 0 to 4 based on degree of cell attachment and proliferation (B).

A

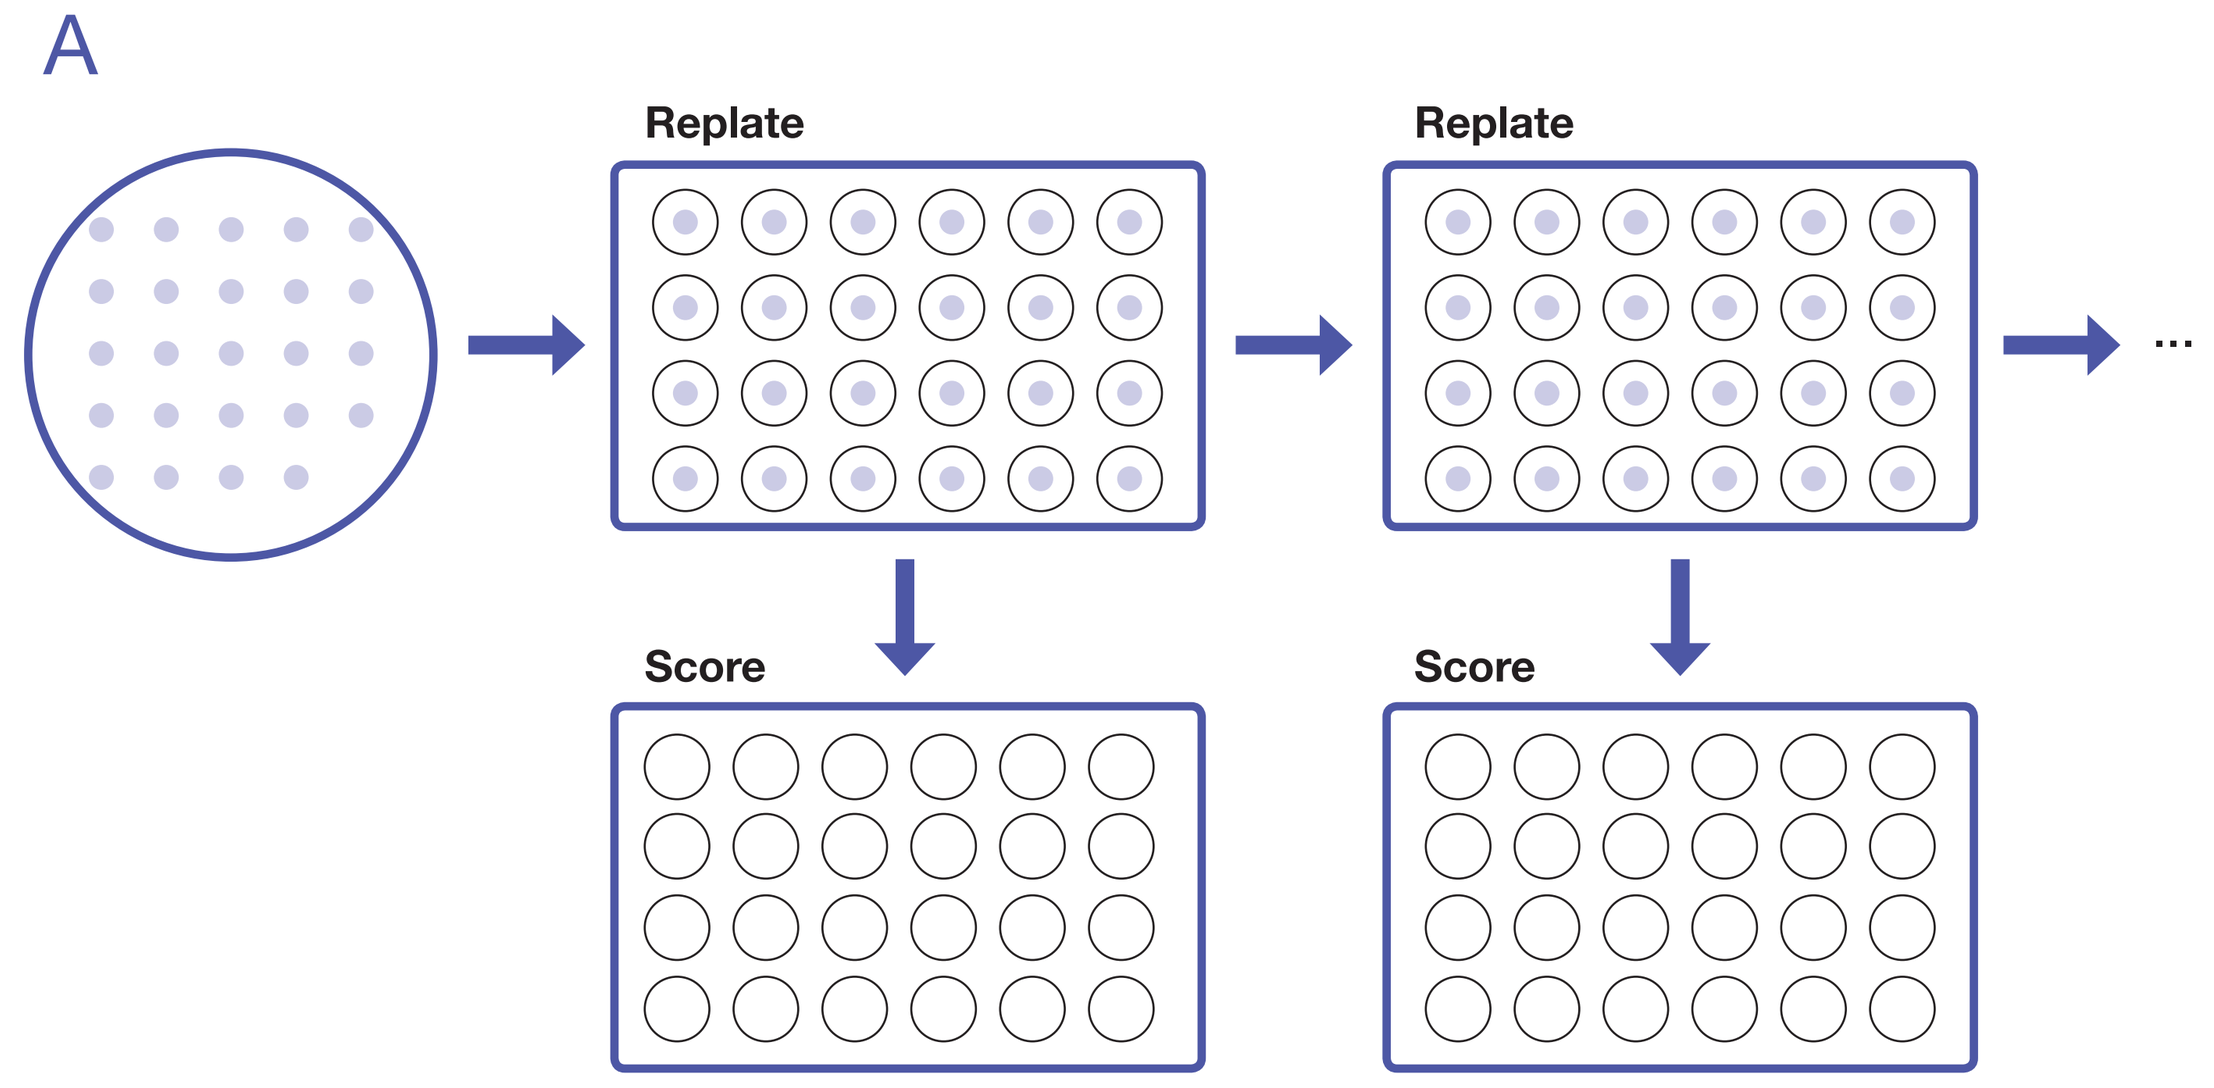
Replate
Replate
Score
Score
...

B

Score	Attachment	Growth
4	Yes	Confluent
3	Yes	Yes
2	Yes	Minimal
1	Minimal	No
0	No	No

100X

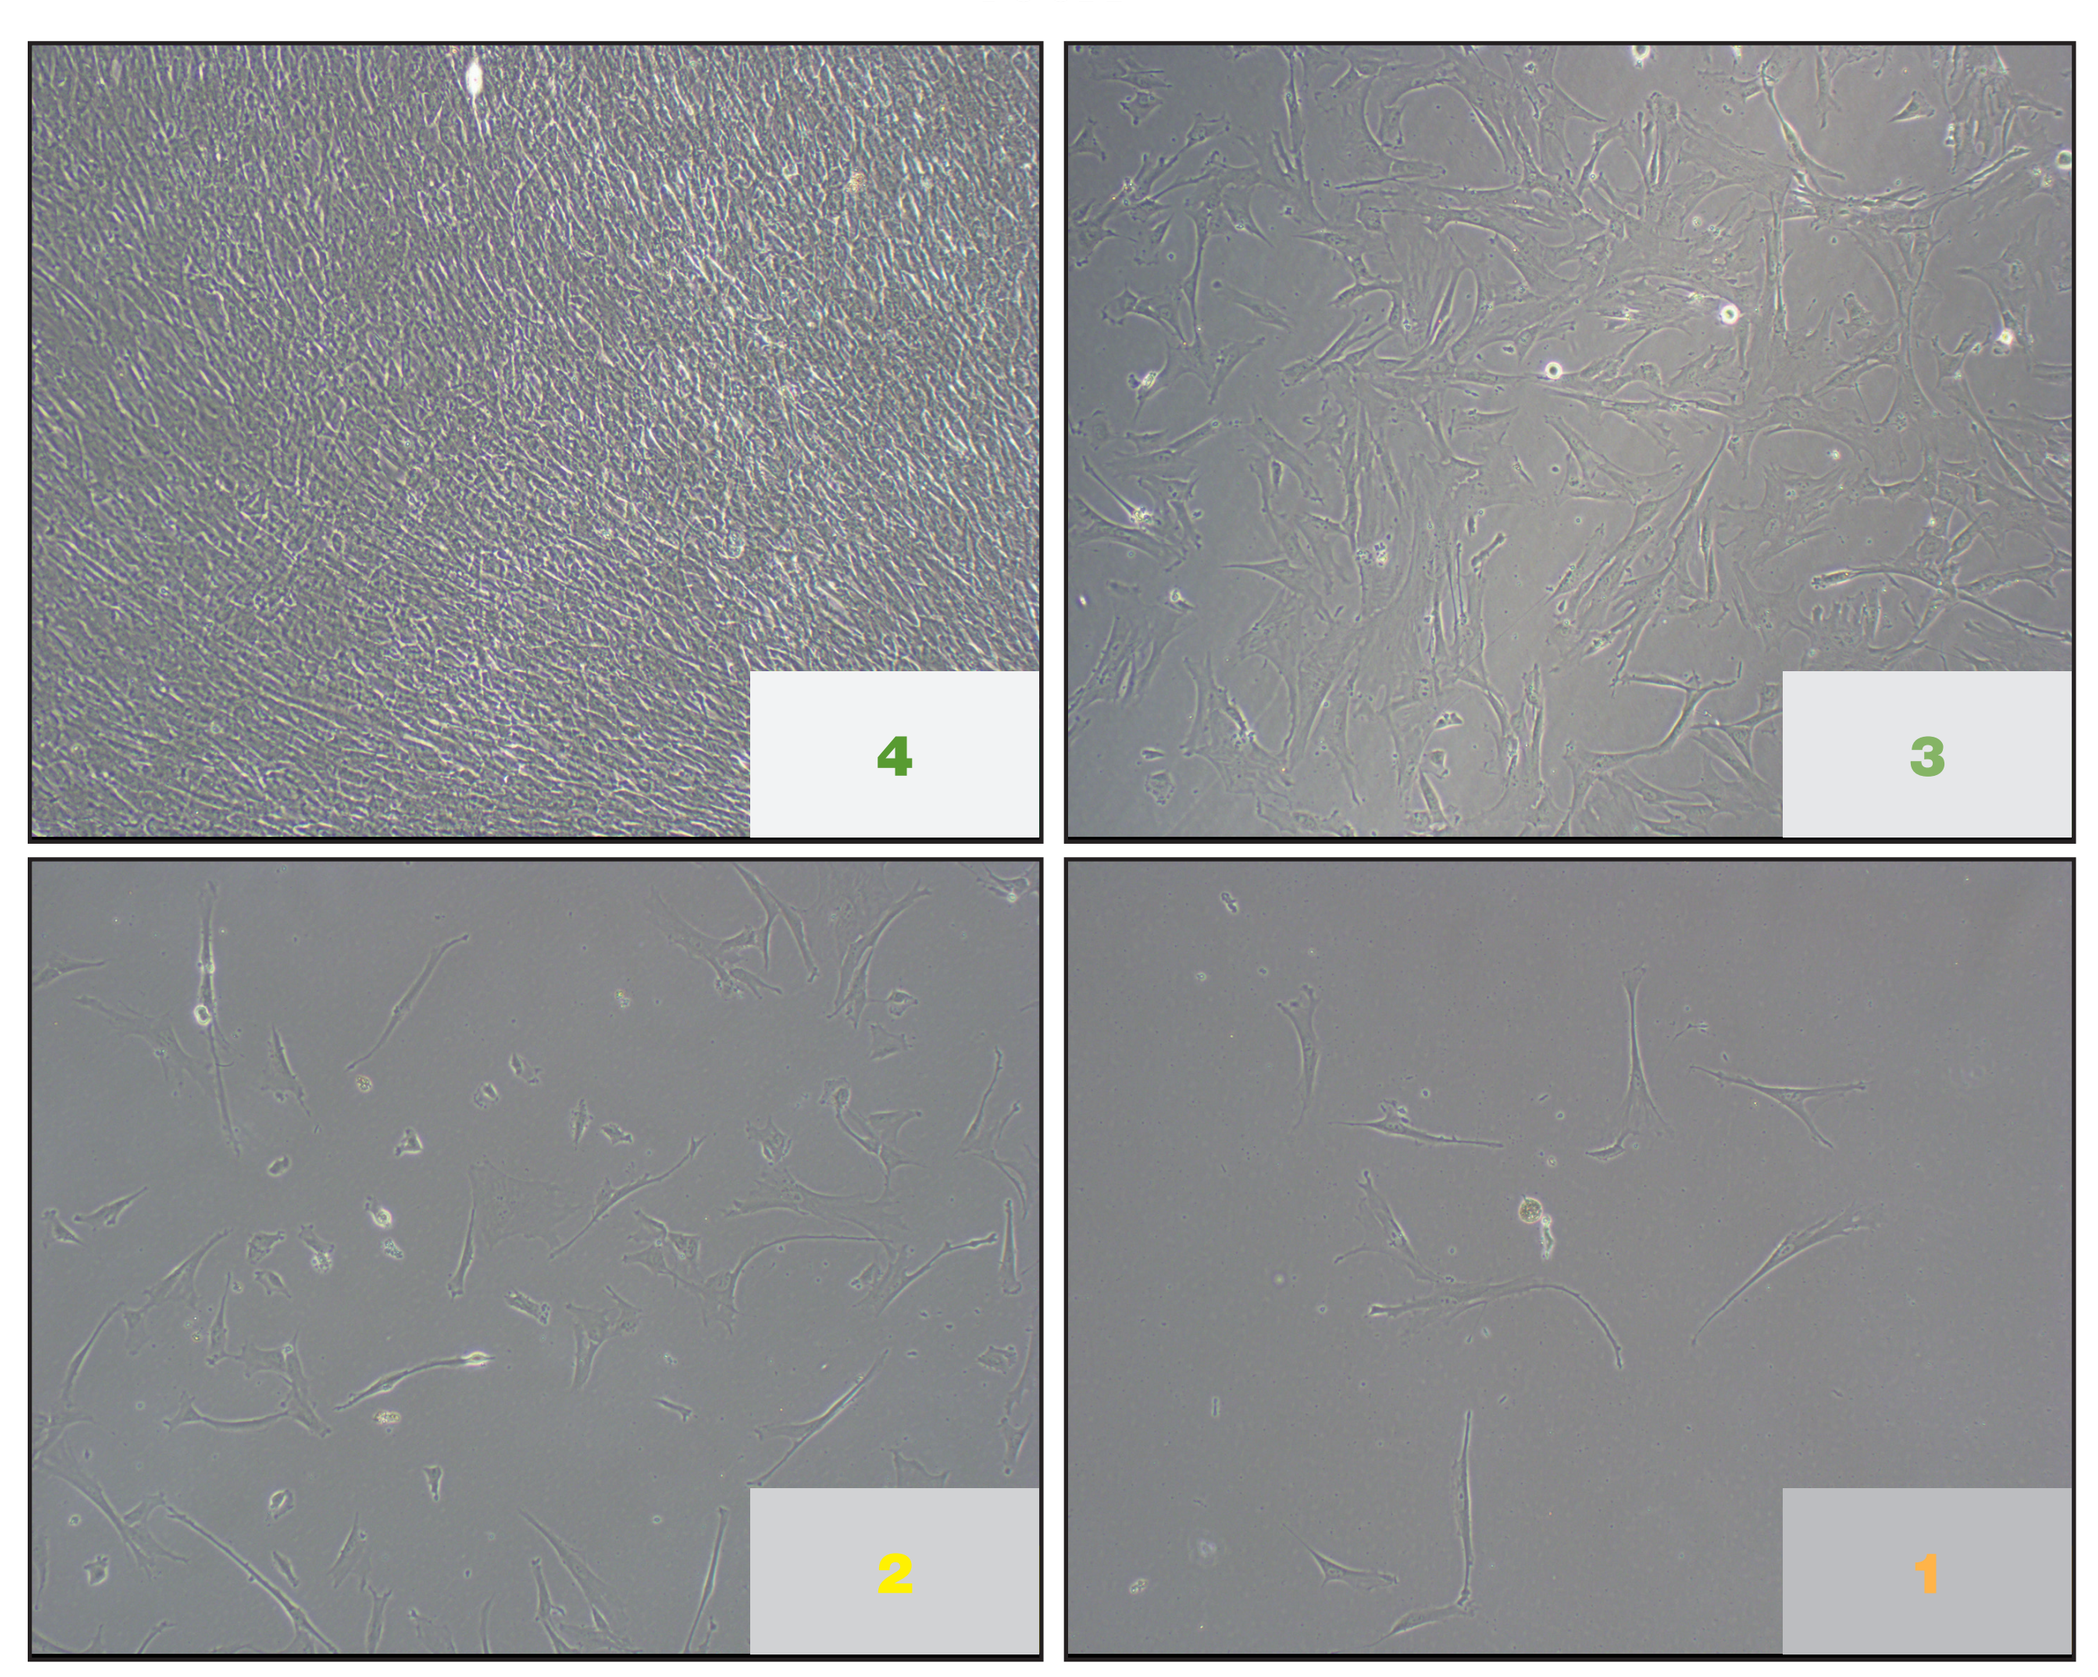
4
3
2
1

Table 1.
Ratio of Explant Scores Compared to the Fresh or Initial Thawed Plating

	Average Score Ratio Compared to Fresh (Std Dev)	p-value	Average Score Ratio Compared to Thawed, Plate 1 (Std Dev)	p-value
Thawed, Plate 1	1.12 (0.28)	>0.05	1 (-)	NA
Replate 1	1.93 (0.53)	0.02*	1.74 (0.21)	0.03*
Replate 2	1.67 (0.51)	>0.05	1.53 (0.46)	>0.05
Replate 3	1.24 (0.59)	>0.05	1.15 (0.51)	>0.05
Replate 4	1.27 (0.74)	>0.05	1.17 (0.67)	>0.05

*Indicates significance ($p < 0.05$)

Figure 2.
Cumulative Fold Increase in Cell Recovery Achieved by Replating CT Explants as Compared to Cell Yield From Initial Thawed Plating

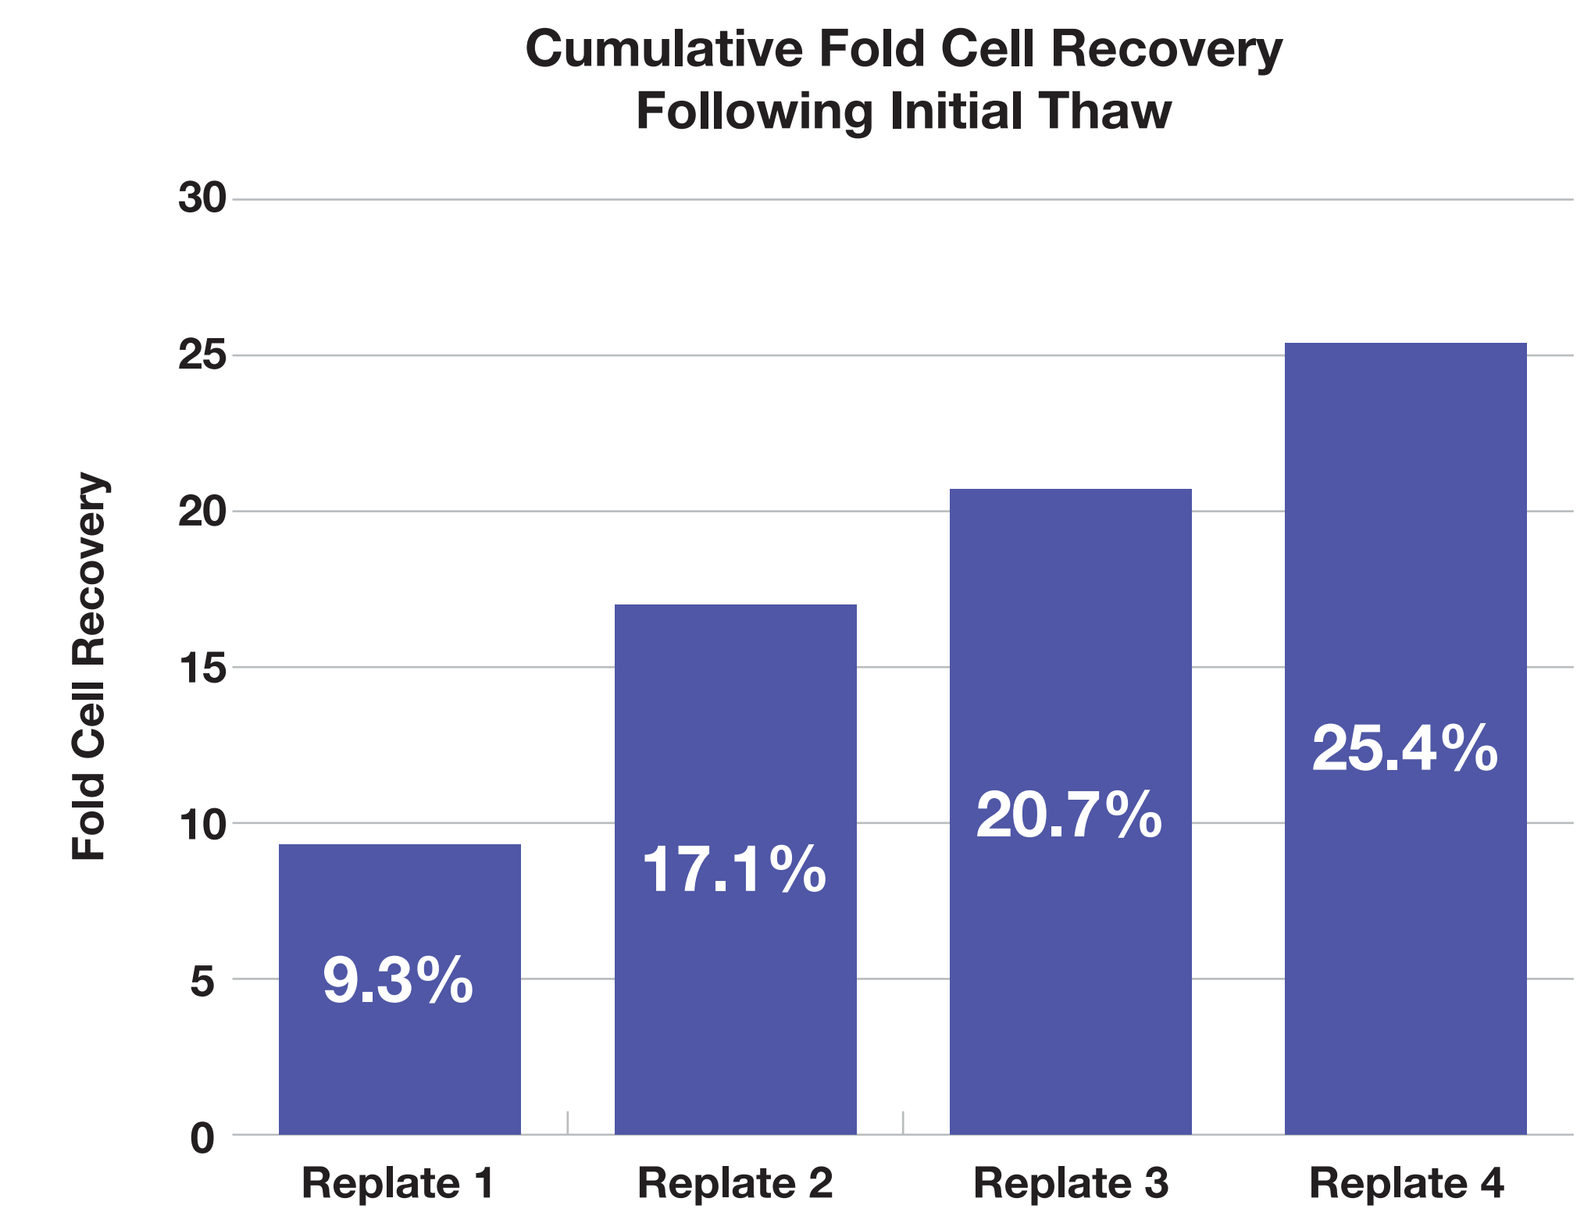
Cumulative Fold Cell Recovery Following Initial Thaw
Fold Cell Recovery
30
25
20
15
10
5
0
9.3%
17.1%
20.7%
25.4%
Replate 1
Replate 2
Replate 3
Replate 4

Table 2.
Average Change in Score for Individual Tissue Pieces Subjected to Serial Explant Plating

Tissue Position	Replate 1 vs. Thawed, Plate 1	Replate 2 vs. Replate 1	Replate 3 vs. Replate 2	Replate 4 vs. Replate 3
1	2	0.75	2	0
2	1.5	0.5	0.5	0.75
3	0.75	0.5	0.75	0.5
4	2	0.75	1.25	1
5	1.25	0	1	0.75
6	1.25	0.75	1.5	0.75
7	2	0.5	1	0
8	0.5	1.25	0.5	0.5
9	2.25	1.25	0.5	0.5
10	1.5	0.5	0.25	1
11	1.25	0.5	0.5	1
12	1.25	0.25	0.75	0.25
13	1.5	0.5	0.75	1.25
14	1.25	0.75	0.5	0.25
15	1	1.25	1.75	2
16	1	0	1	0
17	2.25	0.5	1	0.25
18	2	0.25	0.75	0.25
19	0.5	0.75	0.25	0
20	1	0.5	0.5	0.75
21	0.25	0.25	0.75	0.25
22	1.5	0.5	0.5	0.75
23	1.25	0.25	0.5	0.25
24	1.25	0.25	1	0.75
Average	1.34	0.55	0.82	0.57
Grand Average	0.82			
	—	0.65		

Table 3.
Range of Scores for Individual Tissue Pieces Across Five Serial Explant Platings

Tissue Position	CT Unit 1	CT Unit 2	CT Unit 3	CT Unit 4
1	4	3	3	2
2	1	2	2	3
3	1	2	1	1
4	2	2	3	3
5	2	2	2	2
6	2	1	3	2
7	3	4	1	1
8	1	2	1	2
9	3	3	2	3
10	2	3	1	2
11	1	2	2	1
12	2	1	2	0
13	1	2	4	2
14	1	2	2	2
15	4	4	4	4
16	2	3	1	1
17	4	2	1	4
18	2	3	1	3
19	0	2	2	0
20	2	2	2	1
21	0	2	1	0
22	2	1	2	2
23	1	2	3	0
24	2	2	3	0
Average	1.88	2.25	2.04	1.71
Grand Average	1.97			
Percent of Wells with Minimal Change (Total Range < 3)	79%	71%	71%	75%
Grand Average	74%			